ENHANCED SKIN DEPOSITION OF BETAMETHASONE DIPROPIONATE INTO THE SKIN OF HUMAN VOLUNTEERS FROM CALCIPOTRIENE/BETAMETHASONE DIPROPIONATE CREAM COMPARED TO TOPICAL SUSPENSION

Zoe D. Draelos[1], Matthew M. Draelos[1], Morten Praestegaard[2]

[1]Dermatology Consulting Services, PLLC, High Point, North Carolina; [2]MC2 Therapeutics, Hørsholm, Denmark

INTRODUCTION

- Calcipotriene (CAL) and betamethasone dipropionate (BDP) cream is an effective and convenient treatment of plaque psoriasis made possible by PAD Technology™ [1].
- PAD Technology is a novel innovative topical formulation and drug delivery system that allows formulation and efficient topical delivery of challenging molecules, while at the same time maintaining a favourable aesthetic feel of the formulation.
- The delivery of topical psoriasis medications into the skin is very important for dermatological efficacy. Corticosteroid in combination with calcipotriene is one of the most effective treatments for psoriasis, yet the drug must enter the skin efficiently in order for efficacy to be achieved.[4] One way to improve clinical results is to optimize delivery through enhanced vehicles.

OBJECTIVE

- The objective of the study was to demonstrate the skin deposition of BDP and its major metabolite betamethasone 17-propionate (B17P), from CAL/BDP Cream as compared to CAL/BDP Topical Suspension (TS).

METHODS

- 0.1 ml of CAL/BDP cream and CAL/BDP TS were applied once to four one-inch squares on the forearms of human female volunteers. One one-inch square on each arm served as control with no product applied.
- At 1 hour, 2 hours, 4 hours, and 8 hours after application, tape strips were applied to the application site and held in place for 10 seconds. The first two tape strips removed were discarded to avoid confounding the analysis with residual cream on the skin. This procedure was repeated 20 times on the same area to allow sampling of the stratum corneum (SC) and epidermis down to the level where punctate bleeding occurred indicating intrusion into the dermis. The control square was processed only at 1 hour after application
- Liquid chromatography–mass spectrometry (LCMS) was used to determine the amount of BDP and the metabolite B17P present. Only the odd-numbered tape strips (# 3, 5, 7…19) were analyzed while the even-numbered tape strips (#4, 6, 8…20) were saved as backups.
- A t-test was used to analyze the paired data with statistical significance defined as $p \leq 0.05$.

RESULTS

- The study population included 10 healthy female subjects.
- Consistently higher amounts of BDP were deposited in the skin following CAL/BDP cream application than CAL/BDP TS application at 1 hour, 2 hours, 4 hours, and 8 hours.
- The values in **Figure 1** indicate the total BDP recovered from tape strips (# 3, 5, 7…19) reflecting the total relative amount of drug deposited to SC and epidermis
- Statistical significance was demonstrated in favour of enhanced drug delivery from CAL/BDP cream over CAL/BDP TS ($p \leq 0.05$) at all time points.
- The highest amount of BDP deposited from CAL/BDP cream was present at 1 hour after application with continuing detection at slightly lower levels at hours 2, 4, and 8. BDP deposited from CAL/BDP TS remained lower and consistent at all time points.
- The B17P metabolite was below the lower limit of quantitation in all tested samples.

Figure 1. ng BDP recovered from tape strips reflecting compound deposited to stratum corneum and epidermis

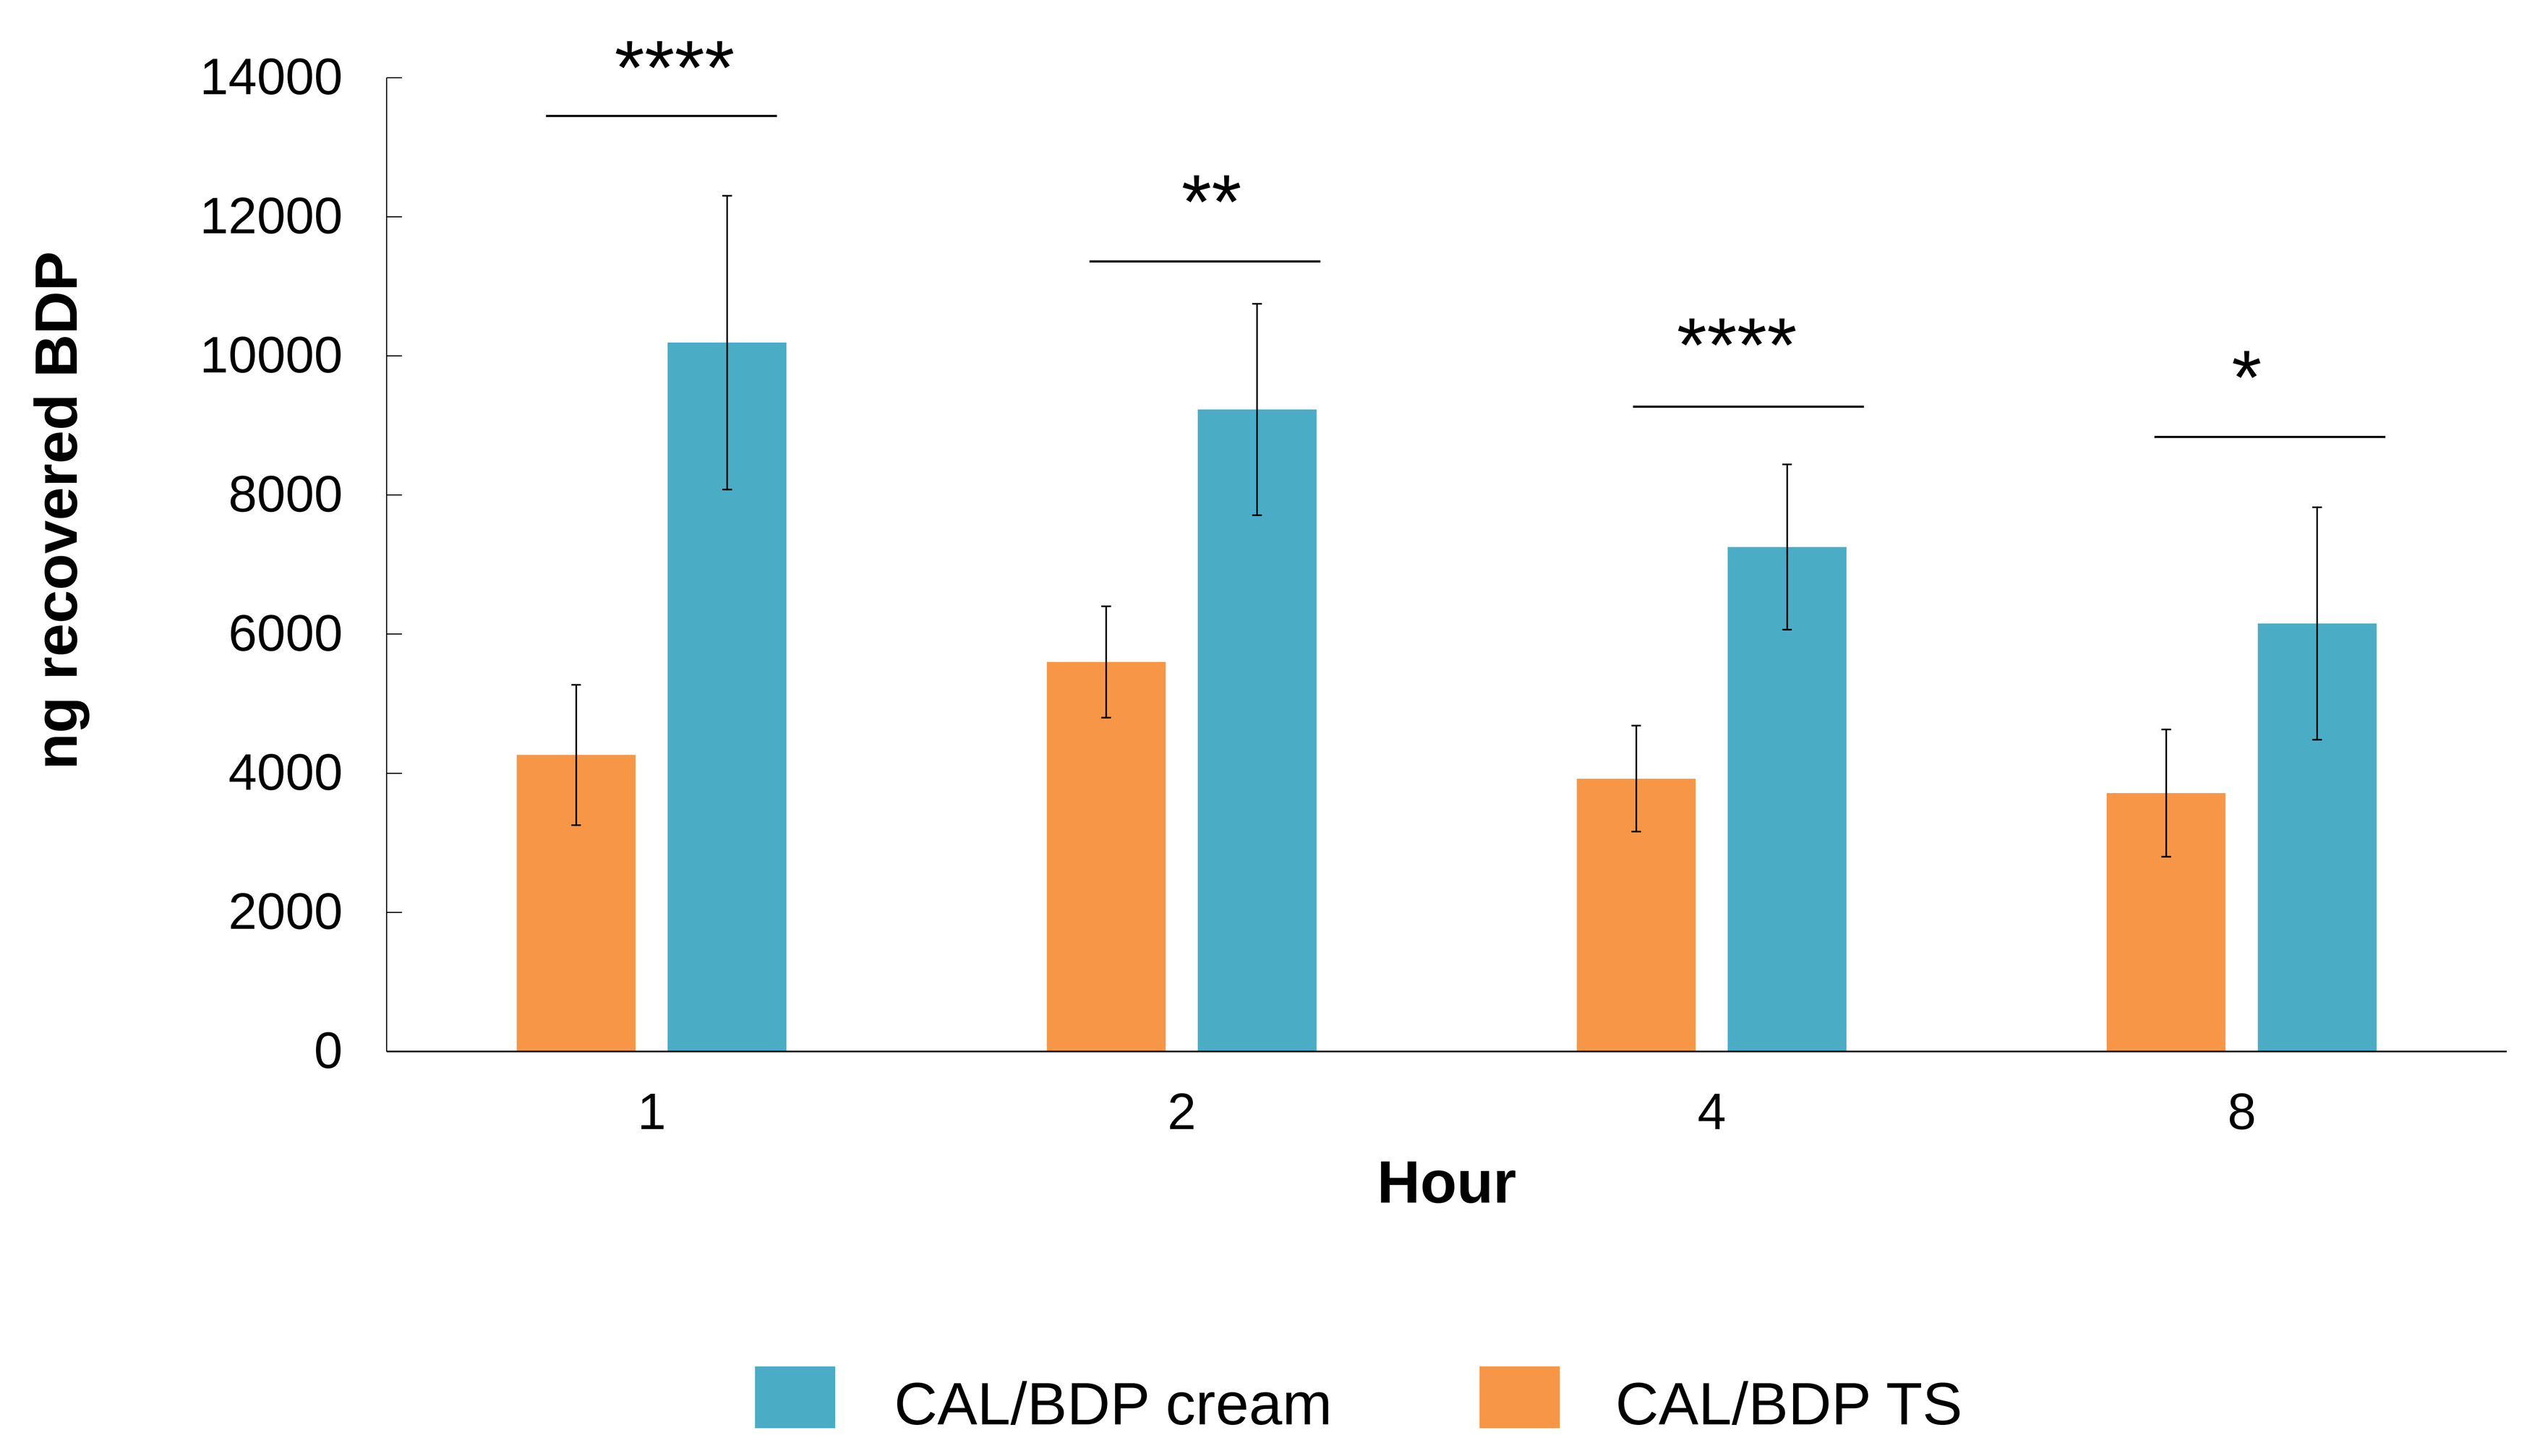
14000
12000
10000
8000
6000
4000
2000
0
ng recovered BDP

**

*
1
2
4
8
Hour
CAL/BDP cream
CAL/BDP TS

Stars denote a statistical significant difference between CAL/BDP cream and TS: *:p<0.05; **: p<0.01; ****:p<0.0001

- In **Figure 2** comparison of BDP recovery is made by drug deposition retrieved from individual tape strips representing progressive skin depth through SC and epidermis.
- Higher amounts of BDP were generally deposited through the skin layers from CAL/BDP cream compared to TS, and as expected, most drug was deposited in the upper skin layers (SC) compared to lowers layers (epidermis) after this single application.

Figure 2. Comparison of recovery of BDP by hours after application from tape strips representing progressive skin depth through SC and epidermis.

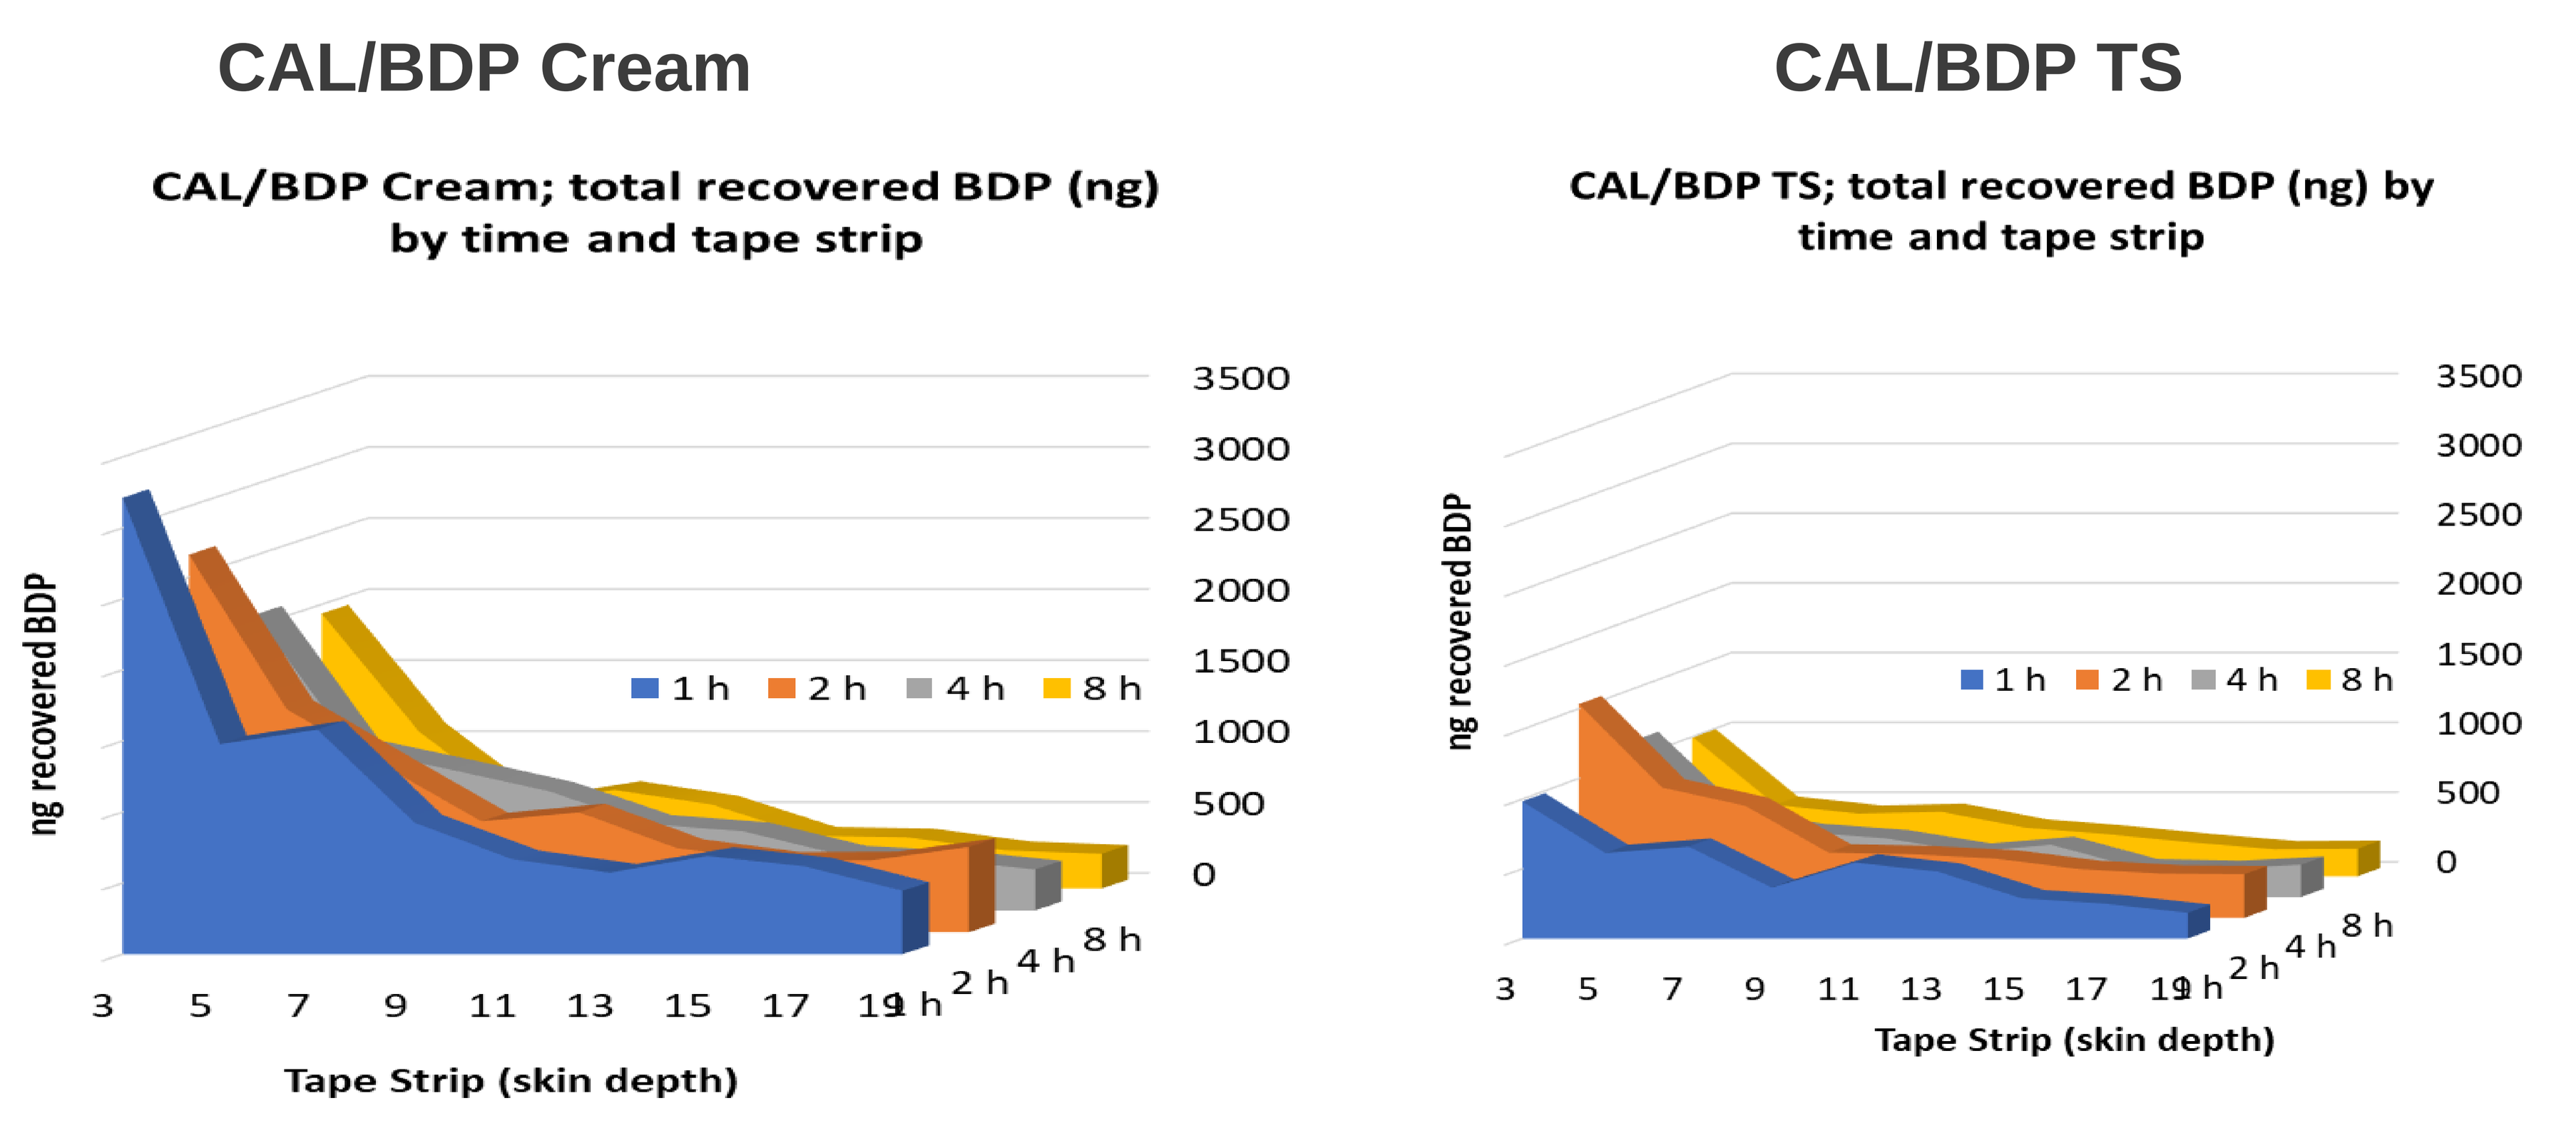
CAL/BDP Cream
CAL/BDP Cream; total recovered BDP (ng) by time and tape strip
ng recovered BDP
1 h
2 h
4 h
8 h
3500
3000
2500
2000
1500
1000
500
0
3 5 7 9 11 13 15 17 19
Tape Strip (skin depth)
CAL/BDP TS
CAL/BDP TS; total recovered BDP (ng) by time and tape strip
ng recovered BDP
1 h
2 h
4 h
8 h
3500
3000
2500
2000
1500
1000
500
0
3 5 7 9 11 13 15 17 19
Tape Strip (skin depth)

CONCLUSION

- CAL/BDP cream is an innovative topical treatment for plaque psoriasis based on PAD™ Technology.
- CAL/BDP cream delivered more BDP into the upper (SC) and lower layers (epidermis) of the skin than CAL/BDP TS. This may account for the observed difference in efficacy in favour of CAL/BDP cream demonstrated in Phase III trials[2].
- This study confirms PAD Technology's benefit in topical delivery in a limited sample size study.
- As a limitation, this study was small and included only female subjects

REFERENCES

1 Praestegaard et al. Dermatology & Therapy, 2022. doi: 10.1007/s13555-022-00794-y

2. Pinter A, et al. JEADV. 2022;36(2):228-36. 3. Wynzora® Cream USPI. 4 Stein Gold et al. J Drugs Dermatol. 2021 Apr 1;20(4):420-425. doi: 10.36849/JDD.2021.5653